EFFECTS OF 8 WEEKS PRE-WORKOUT DIETARY SUPPLEMENT WITH AND WITHOUT SYNEPHRINE ON BLOOD CHEMISTRY PANEL

YP Jung[1], R Dalton[1], C Rasmussen[1], PS Murano[2], CP Earnest[1,3], RB Kreider[1],
Exercise & Sport Nutrition Lab[1], Institute for Obesity Research & Program Evaluation[2],Texas A&M University, College Station, TX.; Nutrabolt[3], Bryan, TX.

Abstract

Background
A number of nutritional strategies have been developed to optimize nutrient delivery prior to exercise. As a result, a number of pre-workout supplements have been developed to increase energy availability, promote vasodilation, and/or positively affect exercise capacity. The purpose of this study was to examine the effects of 8 weeks pre-workout dietary supplement ingestion with and without synephrine on blood chemistry panel.

Methods
In a double-blind, randomized and placebo-controlled manner; 80 apparently healthy and resistance-trained men (21.76±3.59 yr, 15.29±6.19% fat, 25.60±4.03 kg/m^2) ingested in a randomized and counterbalanced manner a dextrose flavored placebo (P); a pre-workout supplement (PWS) containing 3.0 g beta alanine, 2 g creatine nitrate, 2 g arginine AKG, 300 mg N-acetyl tyrosine, 270 mg caffeine, 15 mg Mucuna pruriens; or, the PWS with 20 mg synephrine (PWS+S), and then had blood donation at week 0, week 4, and week 8. The participants had resistance training 4 times per week during 8 weeks supplementation. Data were analyzed by repeated measure MANOVA and presented as means (95% CI) delta change from baseline.

Results
Repeated MANOVA revealed no significant differences among groups in blood urea nitrogen (BUN) (p=0.62) and creatinine (CRE) (p=0.27), and the ratio of BUN/CRE (BCr) (p=0.20). An overall Wilks' Lambda analysis showed significant time effects (p<0.00) in mean changes in BUN (unit conversion to mg/dl by mmol/l x 2.8011) (2.79 mg/dl; 1.58, 4.00) at week 8, CRE (unit conversion to mg/dl by µmol/l x 0.0113) (-0.35 mg/dl; -0.49, -0.21) at week 4 and (-0.16 mg/dl; -0.28, -0.05), and BCr: (8.17; 4.01, 12.33) at week 4 and (7.02; 3.02, 11.02) at week 8. Greenhouse-Geisser univariate analysis revealed no time x group interaction of BUN (p=0.54), CRE (p=0.78), and BCr (p=0.62). In liver enzymes, there were no significant differences among groups in alkaline phosphatase (ALP) (p=0.24), alanine amino transferase (ALT) (p=0.74), and aspartate amino transferase (AST) (p=0.47). Delta analysis revealed significant difference in ALP: (-11.23 U/L; -13.93, -8.5) at week 4 and (-5.44 U/L; -8.48, -2.4) at week 8. LSD Post hoc analysis revealed no significant mean changes in liver enzymes; however, there was a significant difference (p=0.04) of ALP between PWS+S (-3.44 U/L; -6.52, -0.36) and PWS (-7.86 U/L; -10.88, -4.84) compared with P (-5.36 U/L; -8.388, -2.34). However, the range of both groups PWS+S: (68.14±17.39 U/L) at week 4 and (74.44±19.64 U/L) at week 8 and PWS: (87.20±24.72 U/L) at week 4 and (78.49±24.96 U/L) at week 8 were within safe clinical range (30-92 U/L). There were no significant time (p=0.23) and time x group interaction (p=0.78) of creatine kinase (CK) and lactate dehydrogenase (LDH), no significant time x group interaction (p=0.78) of total cholesterol, LDL-C, HDL-C and triglyceride, and a significant time effect (p<0.00) but no time x group effect (p=0.083) of glucose levels.

Conclusion
Ingesting a dietary PWS or PWS+S for 8 weeks had no adverse effect of kidney function, liver enzymes, blood lipid levels, muscle enzymes, and blood sugar levels. These findings are in agreement with other studies testing similar ingredients.

Methods & Procedure

- A double-blind, randomized, and placebo-controlled manner
- 80 apparently healthy and resistance-trained men (21.76±3.59 yr, 15.29±6.19% fat, 25.60±4.03 kg/m^2)
- Supplements
 - Placebo (P): a dextrose flavored
 - Pre-workout (PWS): 3.0 g beta alanine, 2 g creatine nitrate, 2 g arginine AKG, 300 mg of N-acetyl tyrosine, 270 mg caffeine, 15 mg of Mucuna pruriens
 - Pre-workout with synephrine (PWS+S): PWS with 20 mg of synephrine
- At baseline (week 0), blood we drawn, and then at week 4 and week 8, the second and the third blood were drawn.
- Blood chemistry panels were measured at 12 markers by COBAS® C 111 (Roche Diagnostic).
 - Kidney test: Blood urea nitrogen (BUN) and Creatinine (CRE)
 - Liver enzymes: Alkaline phosphatase (ALP), Alanine amino transferase (ALT), and Aspartate amino transferase (AST)
 - Lipid profile: Total cholesterol, HDL-C, LDH -C, an d Triglyceride
 - Muscle inflammation: Creatine kinase (CK), Lactate dehydrogenase (LDH)
 - Glucose

Statistical Analysis

Data were analyzed by repeated measure MANOVA by using IBM SPSS for Windows version 22.0 software (Chicago, IL) and are presented as means (95% CI) delta change from baseline.

Results

- Repeated MANOVA revealed no significant differences among groups in blood urea nitrogen (BUN) (p=0.62) and creatinine (CRE) (p=0.27), and the ratio of BUN/CRE (BCr) (p=0.20).
- There were no significant differences among groups in alkaline phosphatase (ALP) (p=0.24), alanine amino transferase (ALT) (p=0.74), and aspartate amino transferase (AST) (p=0.47).
- There were no significant time (p=0.23) and time x group interaction (p=0.78) of creatine kinase (CK) and lactate dehydrogenase (LDH), no significant time x group interaction (p=0.78) of total cholesterol, LDL-C, HDL-C and triglyceride, and a significant time effect (p<0.00) but no time x group effect (p=0.083) of glucose levels

Conclusions

- Ingesting a dietary PWS or PWS+S for 8 weeks had no adverse effect of kidney function, liver enzymes, blood lipid levels, muscle enzymes, and blood sugar levels.
- These findings are in agreement with other studies testing similar ingredients.

Figures

Kidney Test

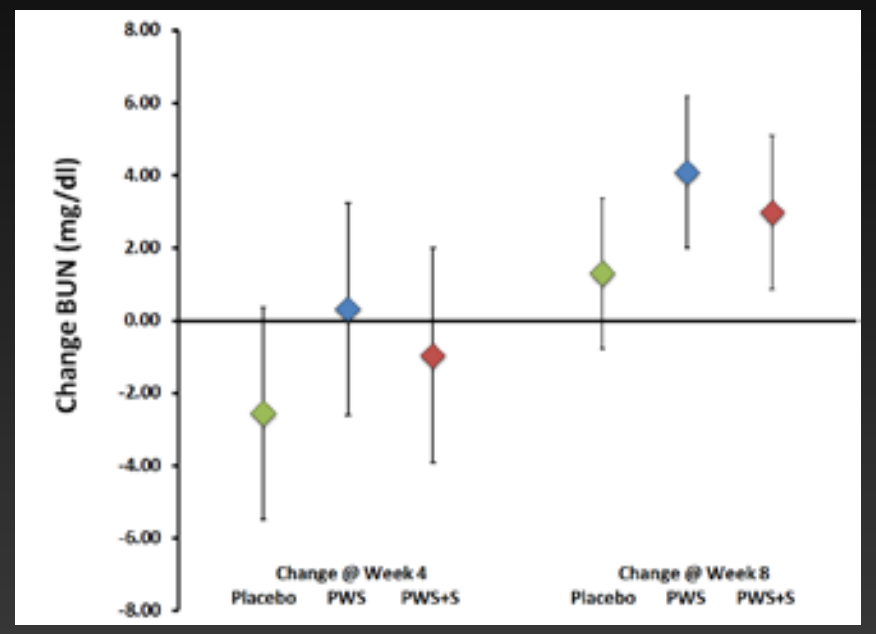
Change BUN (mg/dl)
Change @ Week 4
Placebo PWS PWS+S
Change @ Week 8
Placebo PWS PWS+S

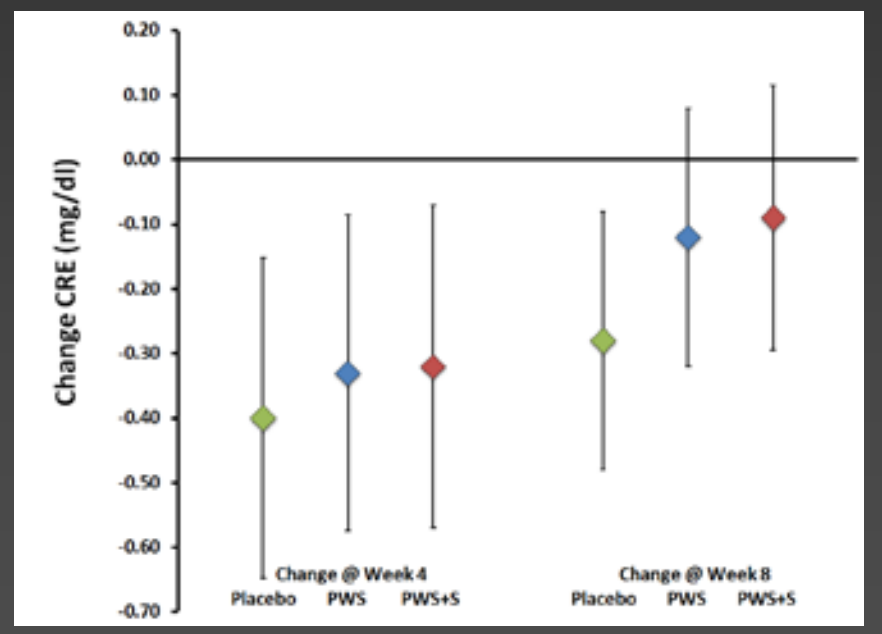
Change CRE (mg/dl)
Change @ Week 4
Placebo PWS PWS+S
Change @ Week 8
Placebo PWS PWS+S

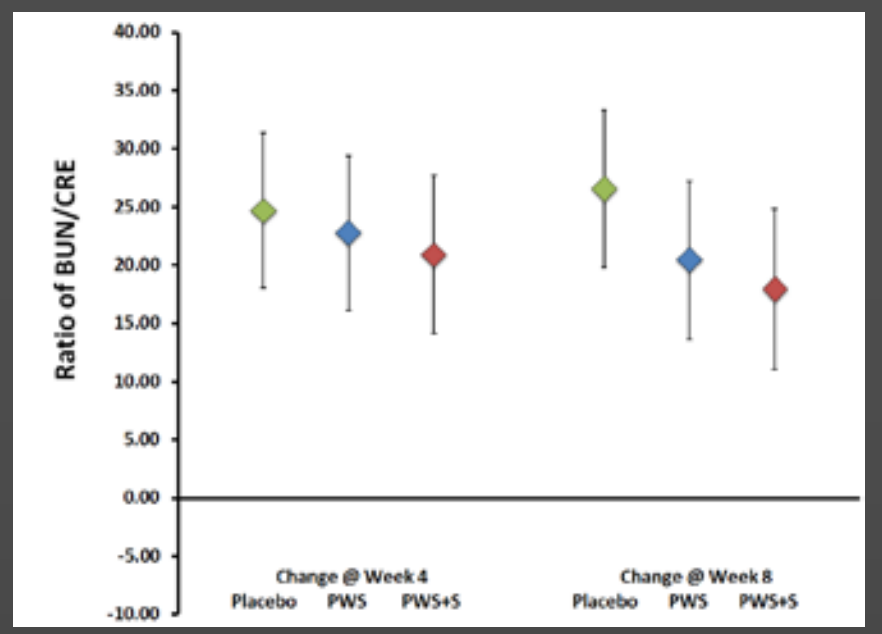
Ratio of BUN/CRE
Change @ Week 4
Placebo PWS PWS+S
Change @ Week 8
Placebo PWS PWS+S

Liver Enzymes

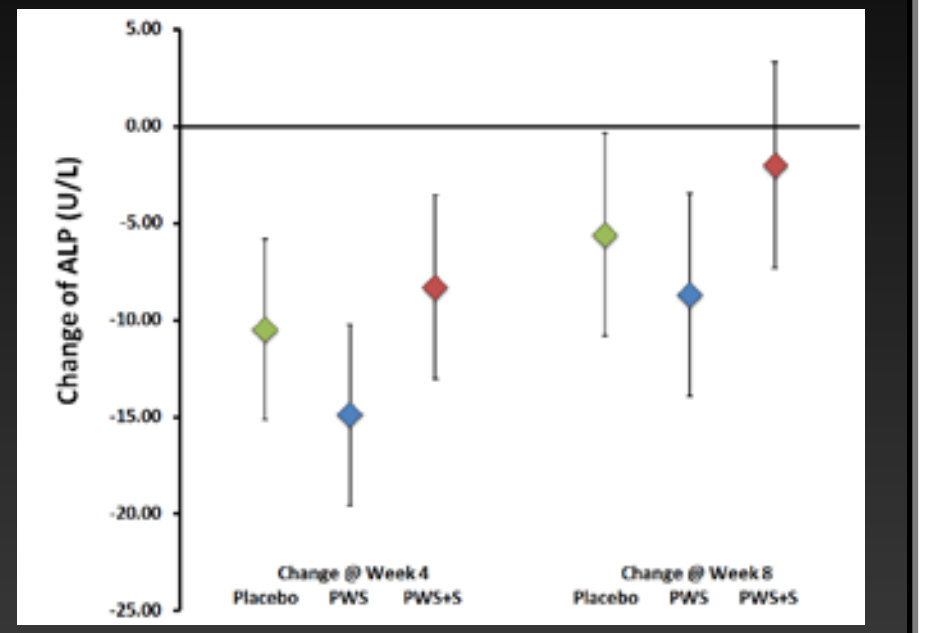
Change of ALP (U/L)
Change @ Week 4
Placebo PWS PWS+S
Change @ Week 8
Placebo PWS PWS+S

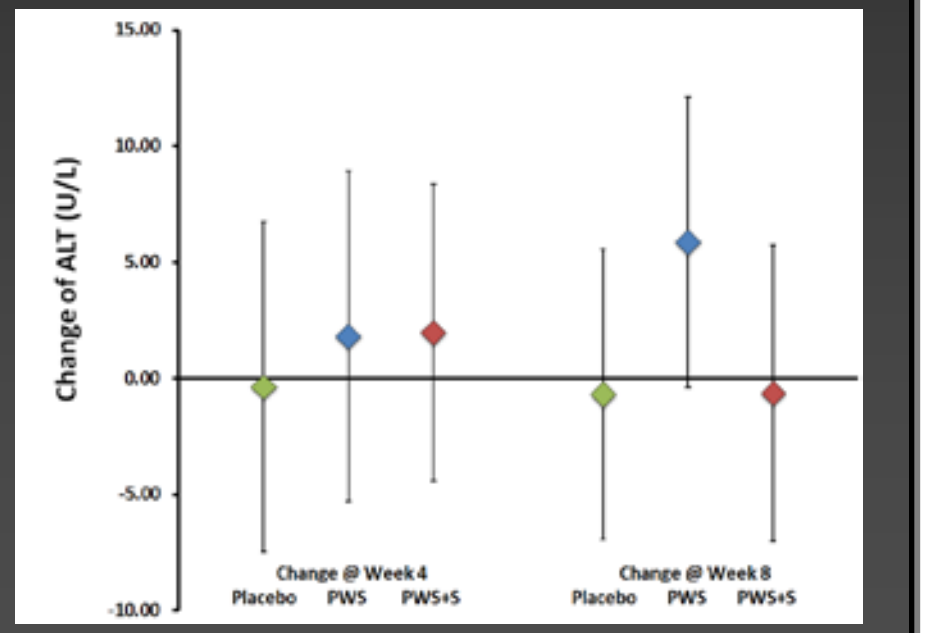
Change of ALT (U/L)
Change @ Week 4
Placebo PWS PWS+S
Change @ Week 8
Placebo PWS PWS+S

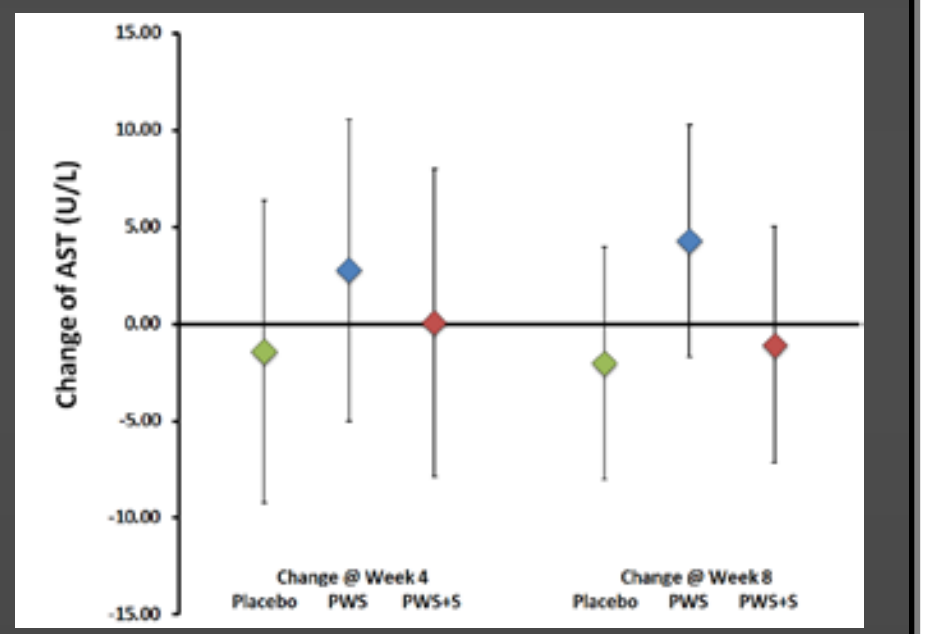
Change of AST (U/L)
Change @ Week 4
Placebo PWS PWS+S
Change @ Week 8
Placebo PWS PWS+S

Muscle Inflammaiton

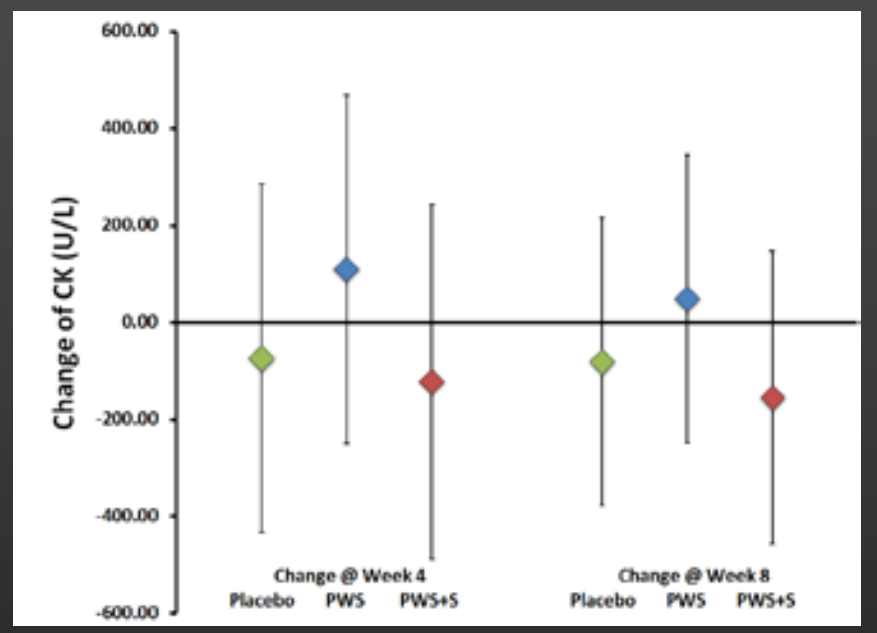
Change of CK (U/L)
Change @ Week 4
Placebo PWS PWS+S
Change @ Week 8
Placebo PWS PWS+S

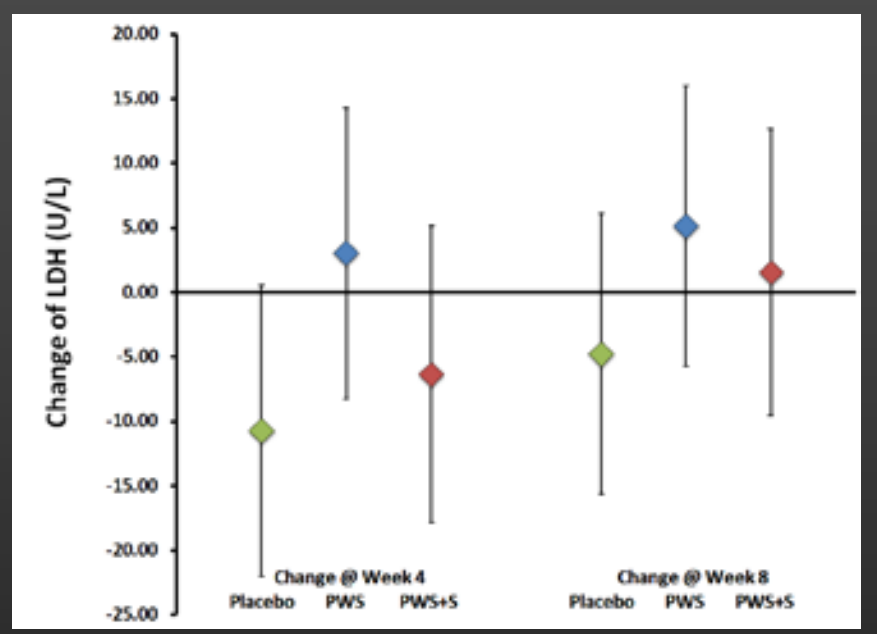
Change of LDH (U/L)
Change @ Week 4
Placebo PWS PWS+S
Change @ Week 8
Placebo PWS PWS+S

Acknowledgements and Disclosures

This study was supported by Nutrabolt through a research grant provided to Texas A&M University. Results do not constitute endorsement of the products studied. CP Earnest serves as Research Director for Nutrabolt and is a Research Associate in the ESNL. RB Kreider serves as a university approved scientific advisor for Nutrabolt. PS Murano serves as quality assurance supervisor.